CS1001, an anti-PD-L1 antibody, combined with Standard-of-Care (SoC) Chemotherapy for First-Line (1L) Advanced GC/GEJ and ESCC: Preliminary Results from 2 Phase 1b Cohorts of CS1001-101 Study

VIRTUAL 2020 ESMO congress

1445P

Lin Shen[1], Jin Li [2], Zhanhui Miao[3], Nong Xu[4], Baorui Liu[5], Xingya Li[6], Qingyuan Zhang[7], Quanli Gao[8], Yanqiu Zhao[9], Hongming Pan[10], Zhidong Pei[11], Wei Li[12] , Hongqiang Xia[13], Jingru Wang[13], Hangjun Dai[13], Qingmei Shi[13], Jason Yang[13]

1.Department of Gastrointestinal Oncology, Key Laboratory of Carcinogenesis and Translational Research (Ministry of Education/Beijing), Peking University Cancer Hospital & Institute 2.Shanghai Eastern Hospital（Oncology） 3.The First Affiliated Hospital of Xinxiang Medical University（Oncology） 4.The First Affiliated Hospital of ZheJiang University（Medical Oncology） 5.Nanjing Drum Tower Hospital（Oncology） 6.The First Affiliated Hospital of Zhengzhou University（Oncology）7.Harbin Medical University Cancer Hospital （Mammary and Lymphatic Medical oncology） 8.Affiliated Cancer Hospital of Zhengzhou University（Biotherapy） 9.Affiliated Cancer Hospital of Zhengzhou University（Medical Oncology） 10.Sir Run Run Shaw Hospital（Medical Oncology） 11.Luoyang Central Hospital（Oncology）12.The First Affiliated Hospital of Jilin University（Oncology）13.CStone Pharmaceuticals (Suzhou) Co., Ltd, Suzhou, China

BACKGROUND

- CS1001 is a high-affinity, full-length, fully human anti-programmed death ligand-1 (PD-L1) immunoglobin G4 (IgG4, s228p) monoclonal antibody developed by OmniRat® transgenic platform which mirrors natural IgG4 human antibody and may potentially reduce the risk of immunogenicity and toxicity in patients (pts).
- In phase 1a of the first-in-human study (NCT03312842), CS1001 at 1200 mg fixed dose every 3 weeks (Q3W) was determined as the recommended phase 2 dose (RP2D) [1]
- Phase 1b is the dose-expansion part of the study to explore the efficacy and safety of CS1001 in multiple cohorts of selected tumor types [2]. Herein, we present the updated efficacy and safety data from 2 cohorts as following:
 - CS1001 in combination with chemotherapy as the first-line (1L) treatment in gastric or gastro-esophageal junction carcinoma (GC/GEJ);
 - CS1001 in combination with chemotherapy as the 1L treatment in esophageal squamous cell carcinoma (ESCC)

METHODS

Key Eligibility:

- Eastern Cooperative Oncology Group (ECOG) performance status of 0-1
- GC/GEJ (1L) cohort: pts with histologically confirmed unresectable locally advanced or metastatic GC/GEJ adenocarcinoma who had not received systemic treatment for advanced or metastatic disease
- ESCC (1L) cohort: pts with histologically confirmed unresectable, locally advanced, recurrent or distantly metastatic ESCC who had not received systemic treatment

Figure 1 - Study Design and Objectives

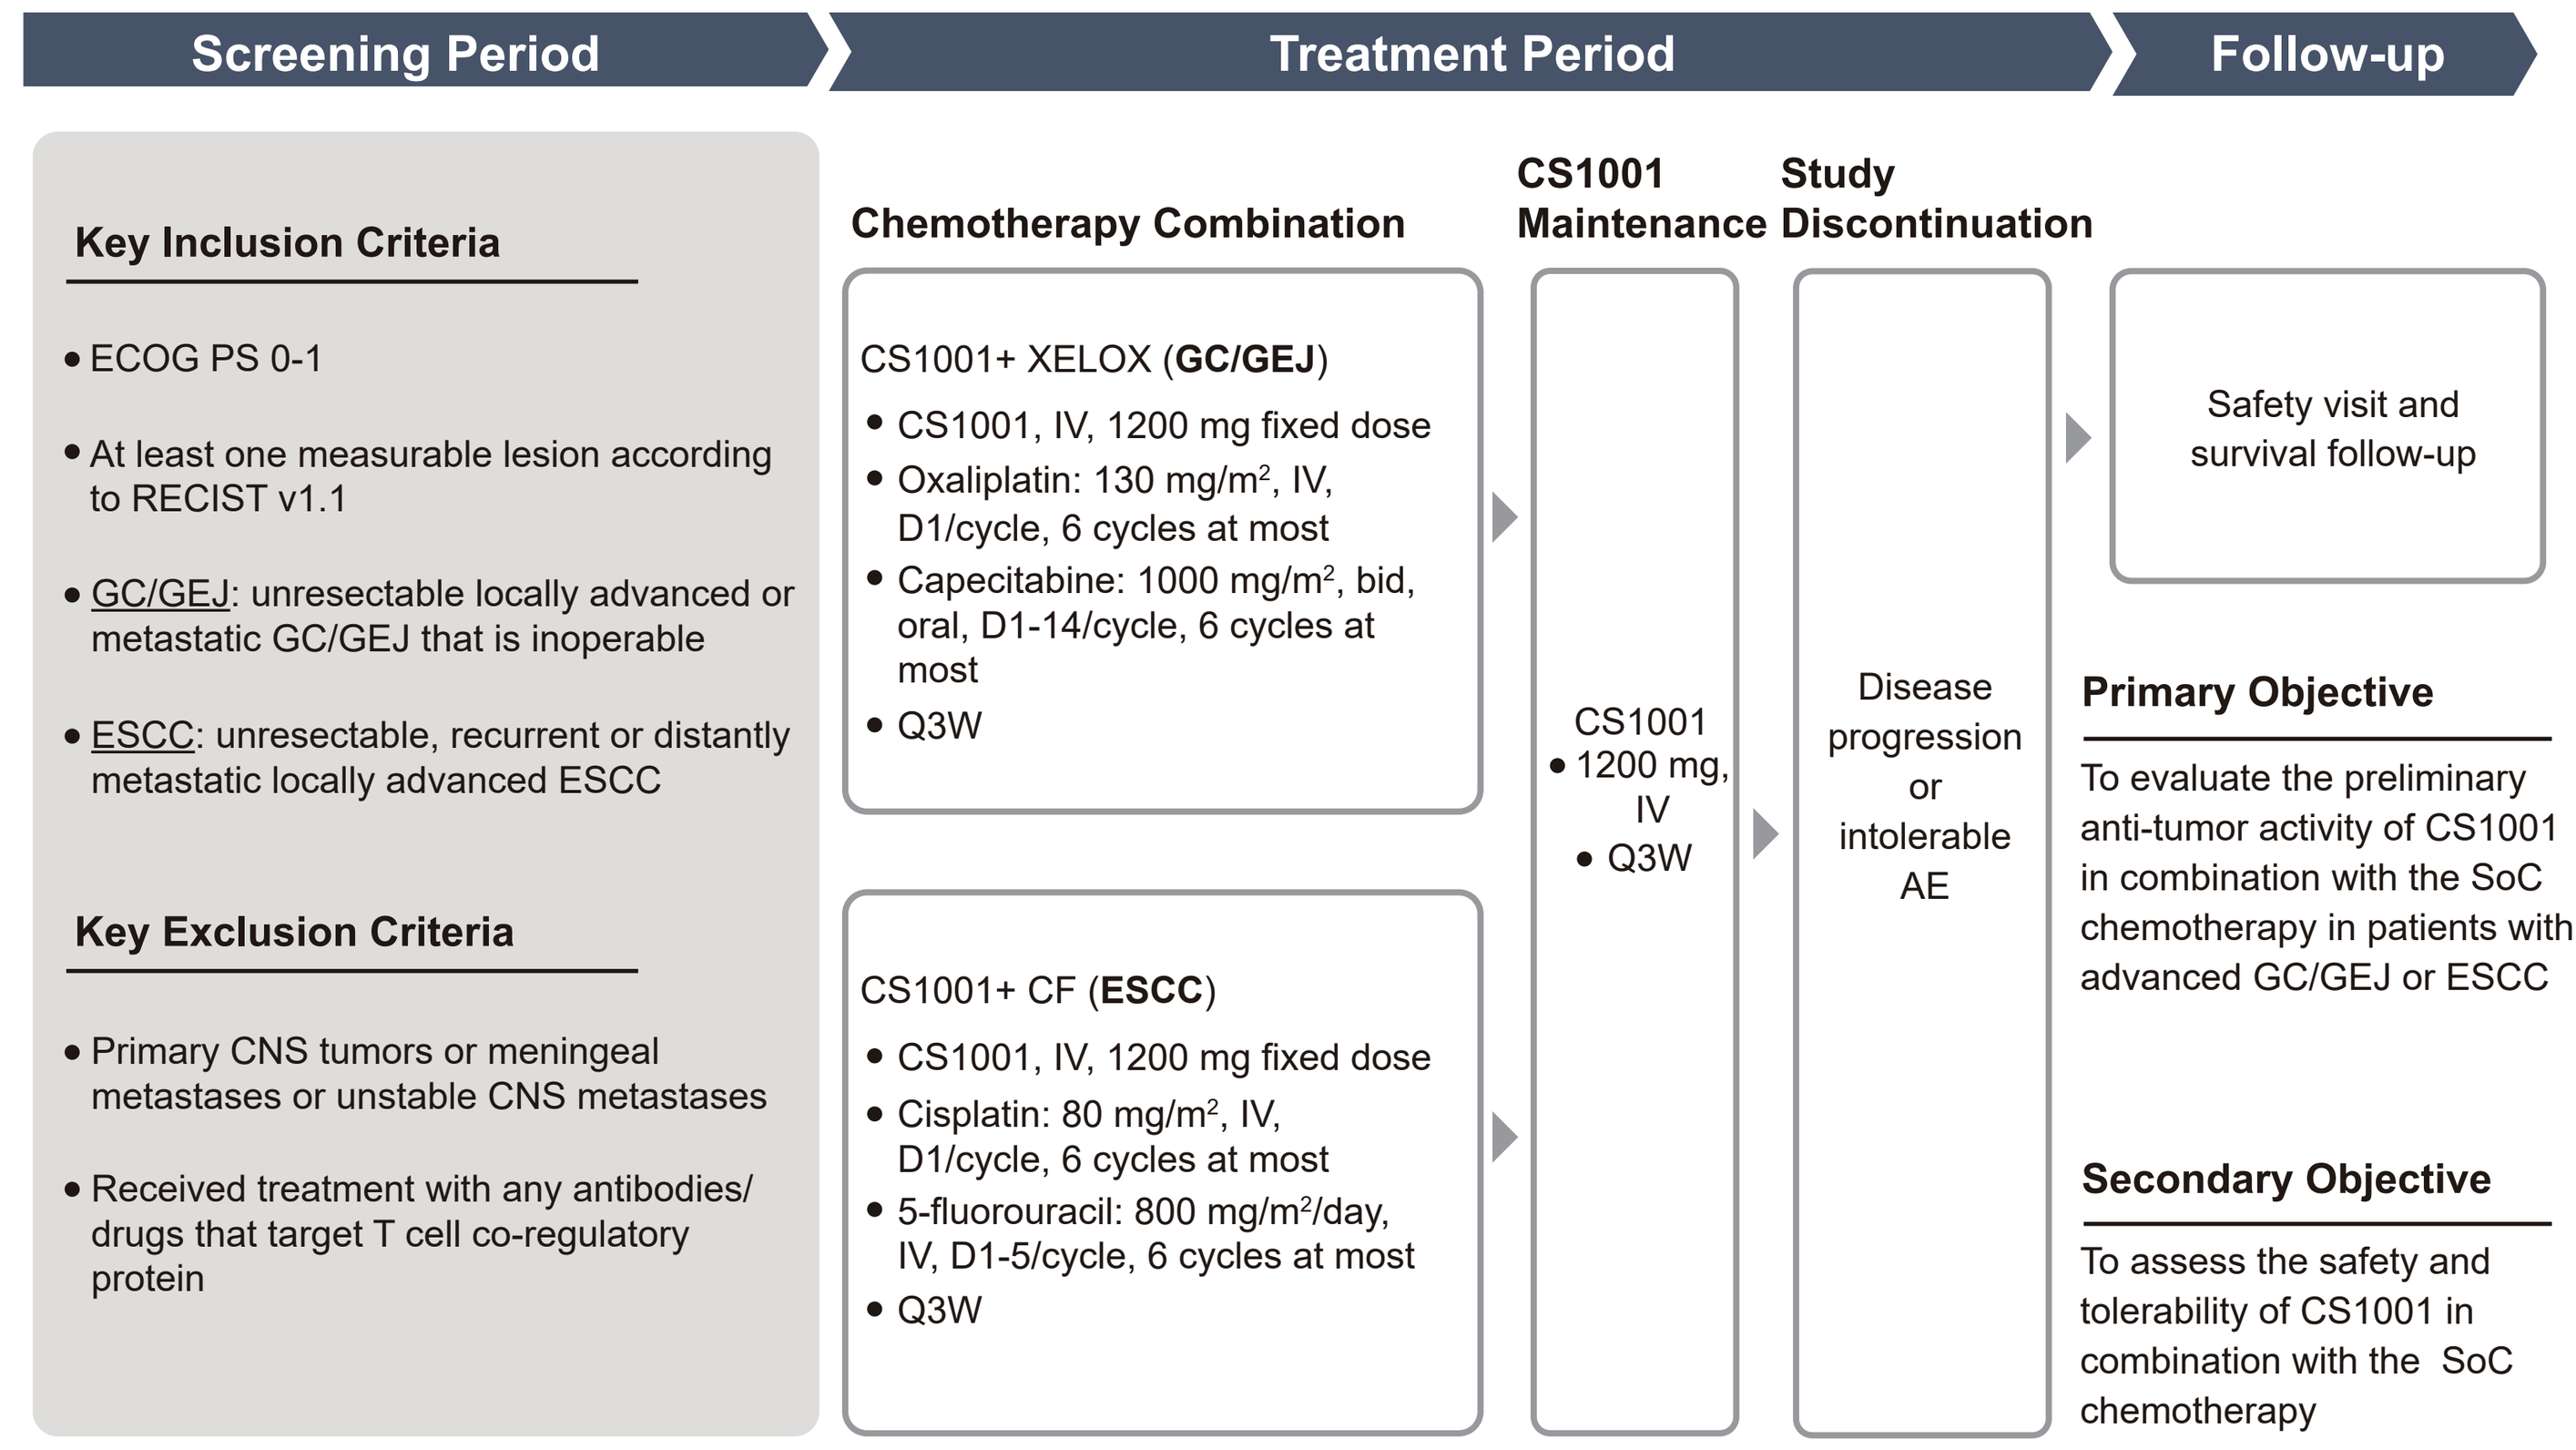
Screening Period
Treatment Period
Follow-up
Key Inclusion Criteria
• ECOG PS 0-1
• At least one measurable lesion according to RECIST v1.1
• GC/GEJ: unresectable locally advanced or metastatic GC/GEJ that is inoperable
• ESCC: unresectable, recurrent or distantly metastatic locally advanced ESCC
Key Exclusion Criteria
• Primary CNS tumors or meningeal metastases or unstable CNS metastases
• Received treatment with any antibodies/ drugs that target T cell co-regulatory protein
Chemotherapy Combination
CS1001+ XELOX (GC/GEJ)
• CS1001, IV, 1200 mg fixed dose
• Oxaliplatin: 130 mg/m², IV, D1/cycle, 6 cycles at most
• Capecitabine: 1000 mg/m², bid, oral, D1-14/cycle, 6 cycles at most
• Q3W
CS1001+ CF (ESCC)
• CS1001, IV, 1200 mg fixed dose
• Cisplatin: 80 mg/m², IV, D1/cycle, 6 cycles at most
• 5-fluorouracil: 800 mg/m²/day, IV, D1-5/cycle, 6 cycles at most
• Q3W
CS1001 Maintenance
CS1001
• 1200 mg, IV
• Q3W
Study Discontinuation
Disease progression or intolerable AE
Safety visit and survival follow-up
Primary Objective
To evaluate the preliminary anti-tumor activity of CS1001 in combination with the SoC chemotherapy in patients with advanced GC/GEJ or ESCC
Secondary Objective
To assess the safety and tolerability of CS1001 in combination with the SoC chemotherapy

AE: Adverse Event; bid: Twice Daily; CNS: Central Nervous System; D: Day; IV: Intravenous; ECOG: Eastern Cooperative Oncology Group; PS: Performance Status; Q3W: Once Every 3 Weeks; SoC: Standard-of-Care

Assessments:

- Tumor assessments were conducted per RECIST v1.1 by investigators every 9 weeks (Q9W) during the first year of treatment and every 12 weeks (Q12W) thereafter
- Adverse events (AEs) were graded according to the National Cancer Institute Common Terminology Criteria for AE (NCI-CTCAE) v4.03

Reference

1. Shen, L., et al., #1165P, Presented at European Society for Medical Oncology (ESMO), 19-23 Oct, 2018, Munich, Germany
2. Shen, L., et al., #1268P, Presented at ESMO, 27 Sep – 1 Oct, 2019, Barcelona, Spain

RESULTS

Patient Disposition, Demographics and Baseline Characteristics (Safety Analysis Set):

- As of 19 Feb 2020, 29 and 39 pts were enrolled in GC/GEJ and ESCC cohorts, respectively
- In the GC/GEJ cohort, 7 pts remained on treatment and 22 discontinued; Reasons of CS1001 discontinuation included progressive disease (n = 13), adverse event (n = 4), patient's withdrawal (n = 3), death (n = 1), and treatment suspension > 6 weeks (n = 1)
- In the ESCC cohort, 16 pts remained on treatment and 23 discontinued; Reasons of CS1001 discontinuation included progressive disease (n = 8), adverse event (n = 5), patient's withdrawal (n = 4), death (n = 2), treatment suspension > 6 weeks (n = 2), protocol deviation (n = 1), and symptomatic deterioration (n = 1).

Table 1 - Demographics and Baseline Characteristics (Safety Analysis Set)

Characteristics		GC/GEJ	ESCC
		N = 29	N = 39
Age (years)	Median (range)	60 (40, 73)	61 (45, 75)
Sex, n (%)	Male	23 (79.3)	29 (74.4)
	Female	6 (20.7)	10 (25.6)
ECOG PS, n (%)	0	12 (41.4)	8 (20.5)
	1	17 (58.6)	31 (79.5)
Initial diagnosis	n (%)	GC: 26 (89.7) GEJ: 3 (10.3)	ESCC: 39 (100)
Time since initial diagnosis (month)	Median (range)	0.9 (0.12, 79.2)	4.9 (0.12, 36.5)
Prior anti-cancer therapy regimen	Median (range)	0 (0, 2)*	0 (0, 2)**
Current cancer stage, n (%)	Stage III	1 (3.4)	4 (10.3)
	Stage IV	28 (96.6)	35 (89.7)

ECOG – Eastern Cooperative Oncology Group; PS – Performance Status

* A total of 10 patients received neoadjuvant and/or adjuvant therapy, among which 1 patient received both neoadjuvant and adjuvant therapies around curative surgery

** A total of 8 patients received neoadjuvant and/or adjuvant therapy, among which 1 patient received cisplatin + paclitaxel after radical surgery and later changed to cisplatin + capecitabine due to allergy to taxol

Efficacy:

- As of 19 Feb 2020, all 29 pts from GC/GEJ cohort were included in the efficacy analysis set. Eighteen (62.1%) pts achieved partial response (PR), including 17 confirmed PRs and 1 unconfirmed PR; 6 (20.7%) pts had stable disease (SD); 3 (10.3%) pts experienced progressive disease (PD); 2 (6.9%) pts discontinued without having any post-baseline tumor assessment (i.e. NA)
- As of 19 Feb 2020, 37 pts from the ESCC cohort were included in the efficacy analysis set. Two pts were excluded because both were on treatment and had not yet reached the time for the 1st post-baseline tumor assessment. Twenty-five (67.6%) pts achieved PR, including 20 confirmed PRs and 5 unconfirmed PRs; 8 (21.6%) pts had SD; 2 (5.4%) pts experienced PD; 2 (5.4%) pts discontinued without having any post-baseline tumor assessment (i.e. NA)

Table 2 - Summary of Objective Response, PFS, OS (Efficacy Analysis Set)

	GC/GEJ (N = 29)	ESCC (N = 37)
ORR, n (%)	18 (62.1)	25 (67.6)
Best overall response, n (%)		
PR	18 (62.1)	25 (67.6)
SD	6 (20.7)	8 (21.6)
PD	3 (10.3)	2 (5.4)
NA	2 (6.9)	2 (5.4)
DCR, n (%)	24 (82.8)	33 (89.2)
Median DoR (month, range)	11.3 (1.0+, 14.1+)	NR (0.03+, 13.3+)
6-month DoR rate (%, 95% CI)	76.5 (48.8, 90.4)	80.2 (50.1, 93.2)
Median PFS (month, range)	8.3 (1.4, 16.1+)	9.0 (2.0, 15.2+)
Median OS (month, range)	17.0 (1.4, 18.7+)	NR (2.5, 18.2+)

NA – Not Applicable; NR – Not Reached; ORR – Objective Response Rate; DCR – Disease Control Rate; DoR – Duration of Response; PFS – Progression-Free Survival; OS – Overall Survival

"+" stands for the values of censored patients

Figure 2 – Waterfall Plot of Maximum Target-Lesion Shrinkage by RECIST v1.1 (Efficacy Analysis Set)

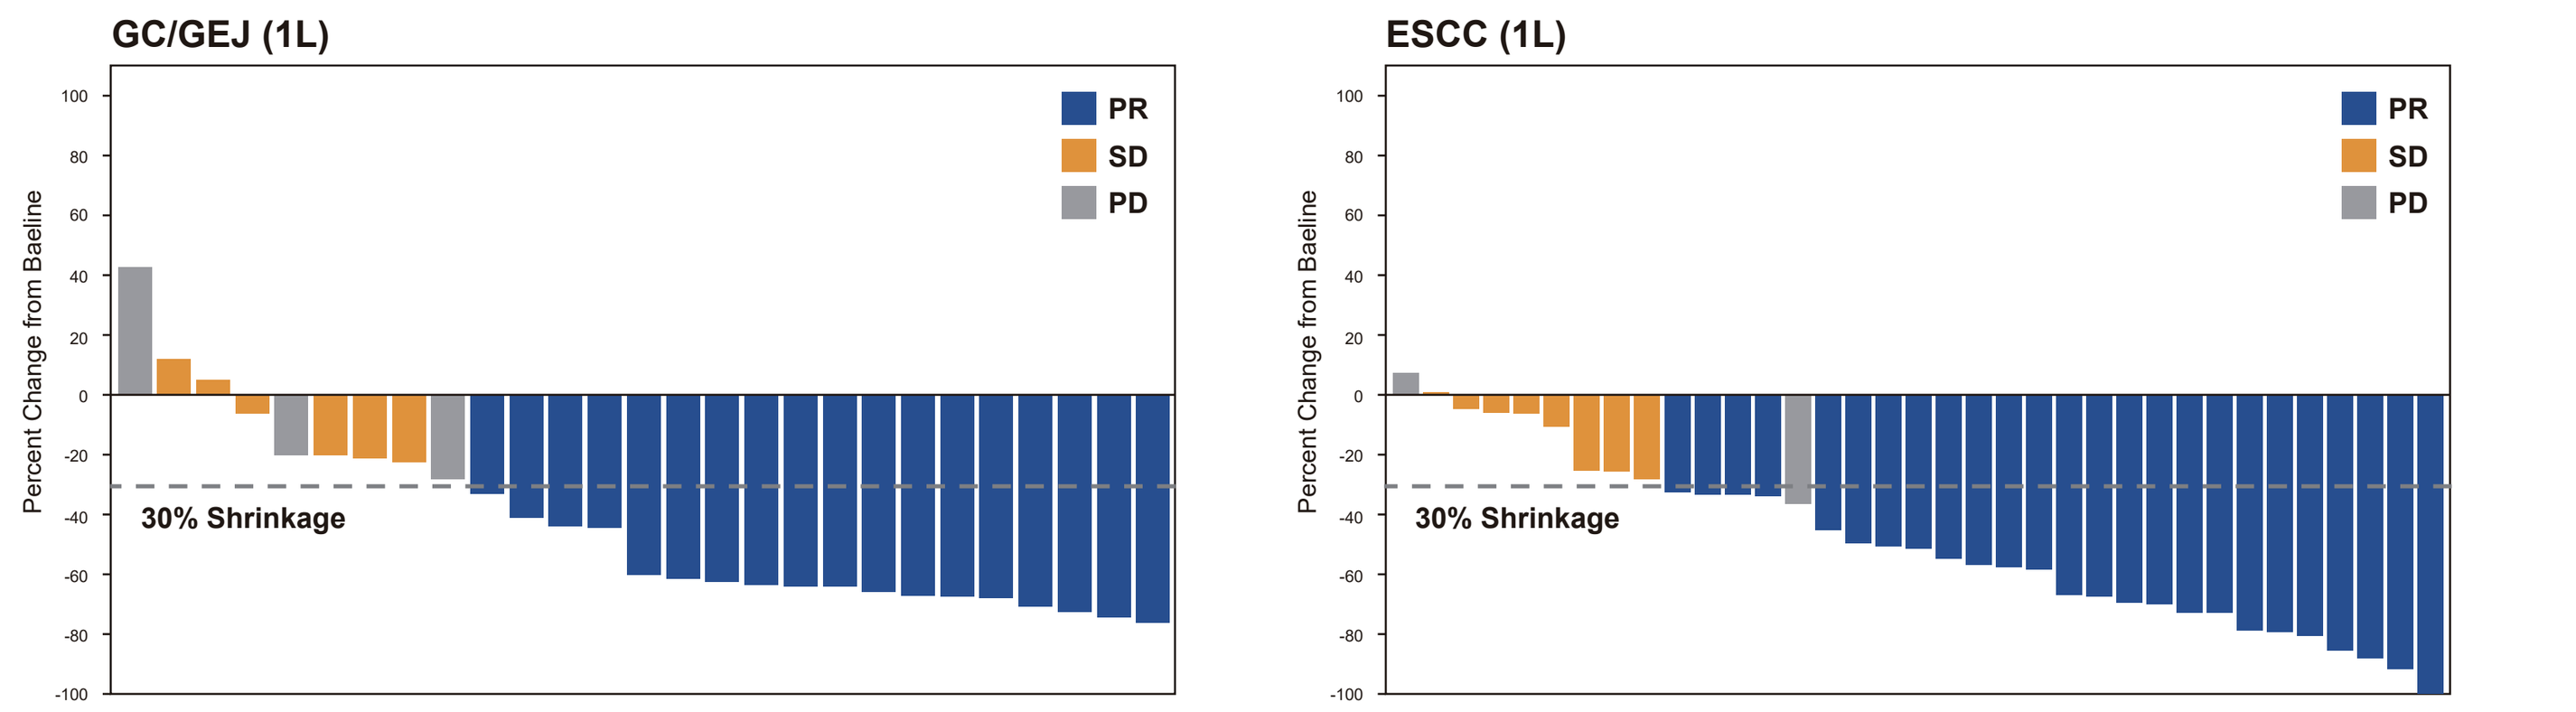
GC/GEJ (1L)
ESCC (1L)
Percent Change from Baseline
30% Shrinkage
PR
SD
PD

Note: 2 patients from each of GC/GEJ and ESCC cohorts were not shown due to lack of post-baseline tumor assessment

Figure 3 – Swimmer Plot of Treatment Duration and Tumor Assessment by RECIST v1.1 (Efficacy Analysis Set)

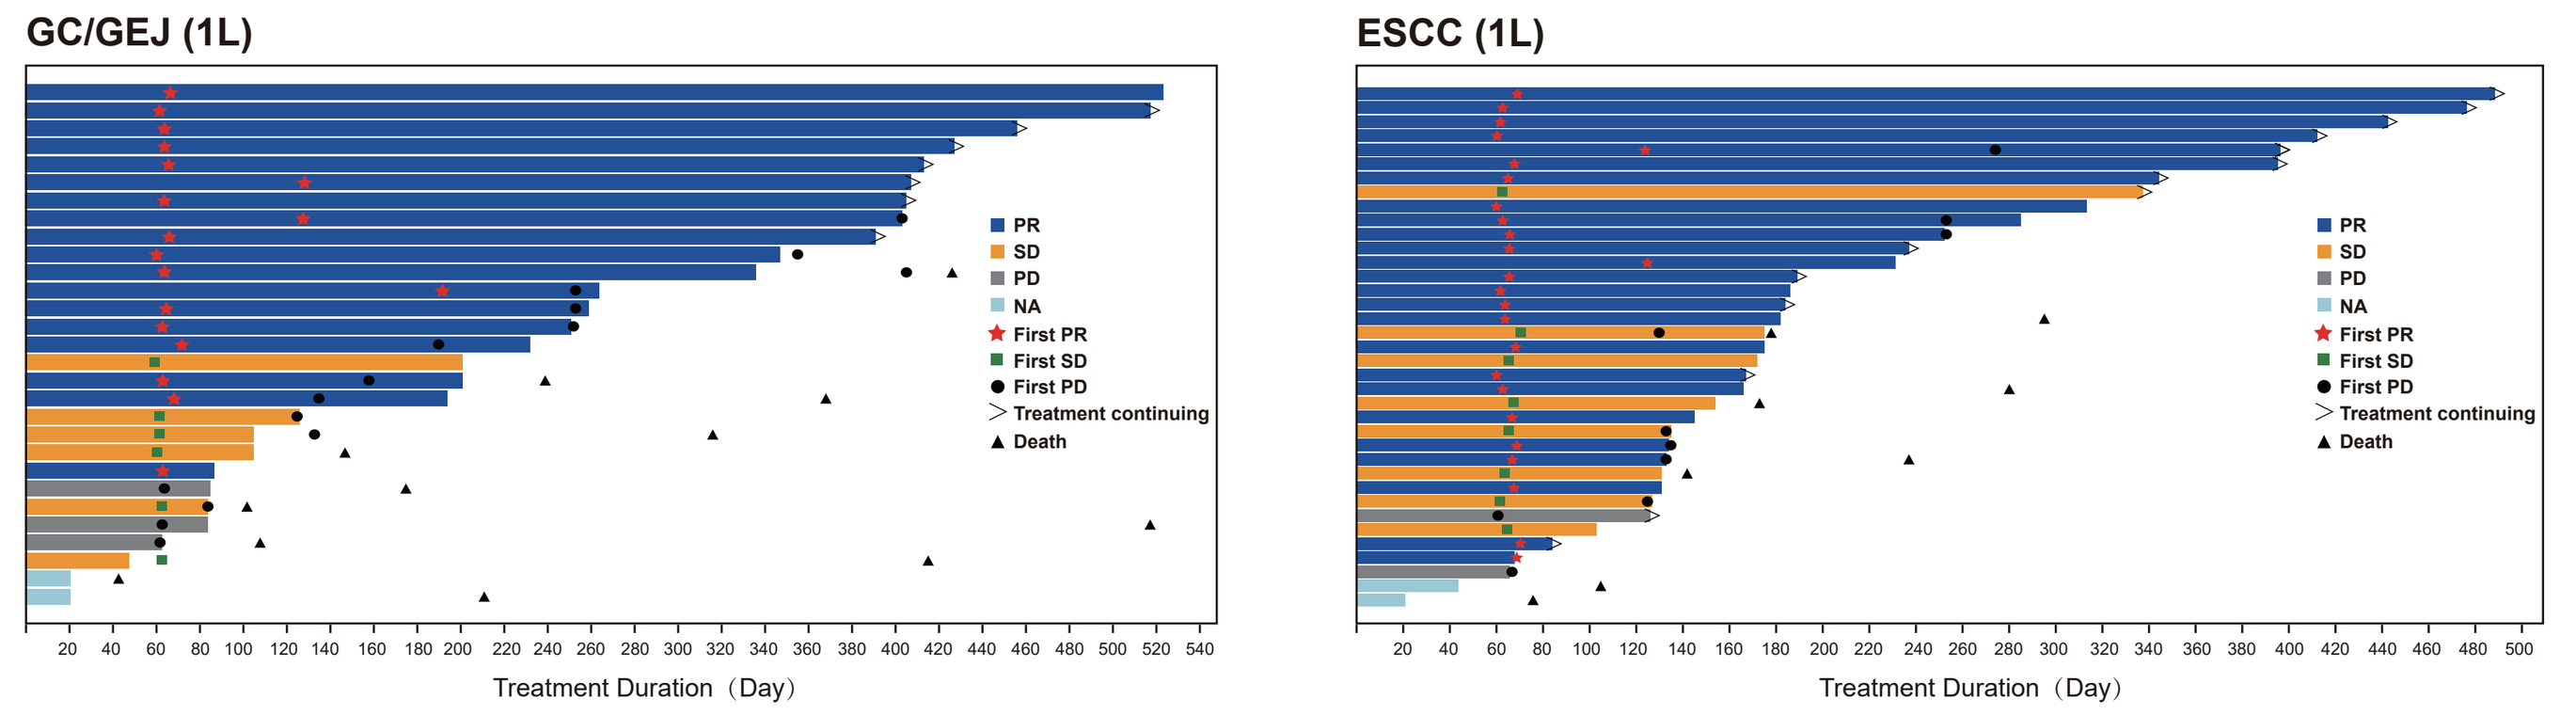
GC/GEJ (1L)
ESCC (1L)
PR
SD
PD
NA
First PR
First SD
First PD
Treatment continuing
Death
Treatment Duration（Day）

Figure 4 – Spider Plot of Percentage Change from Baseline in the Sum of Diameters (Efficacy Analysis Set)

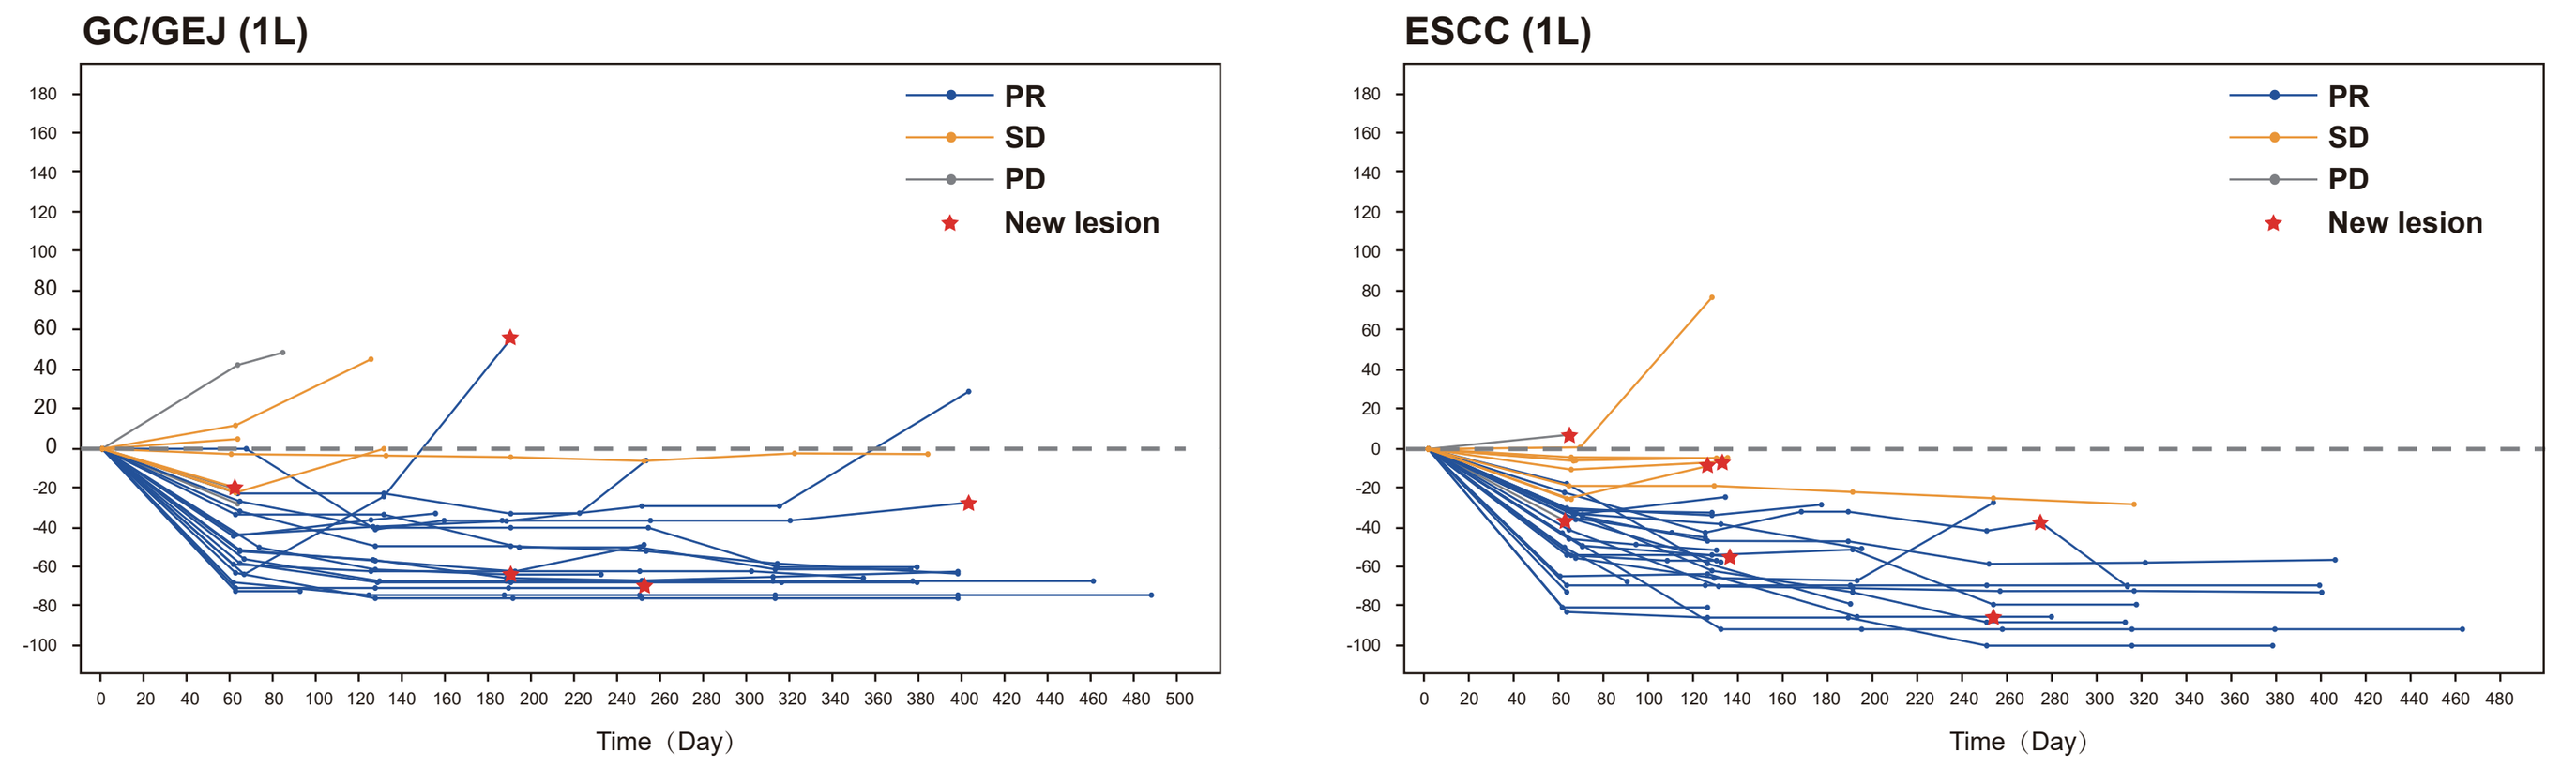
GC/GEJ (1L)
ESCC (1L)
PR
SD
PD
New lesion
Time（Day）

Note: 2 patients from each of GC/GEJ and ESCC cohorts were not shown due to lack of post-baseline tumor assessment

Figure 5 – Representative CT Scan Images of Responders

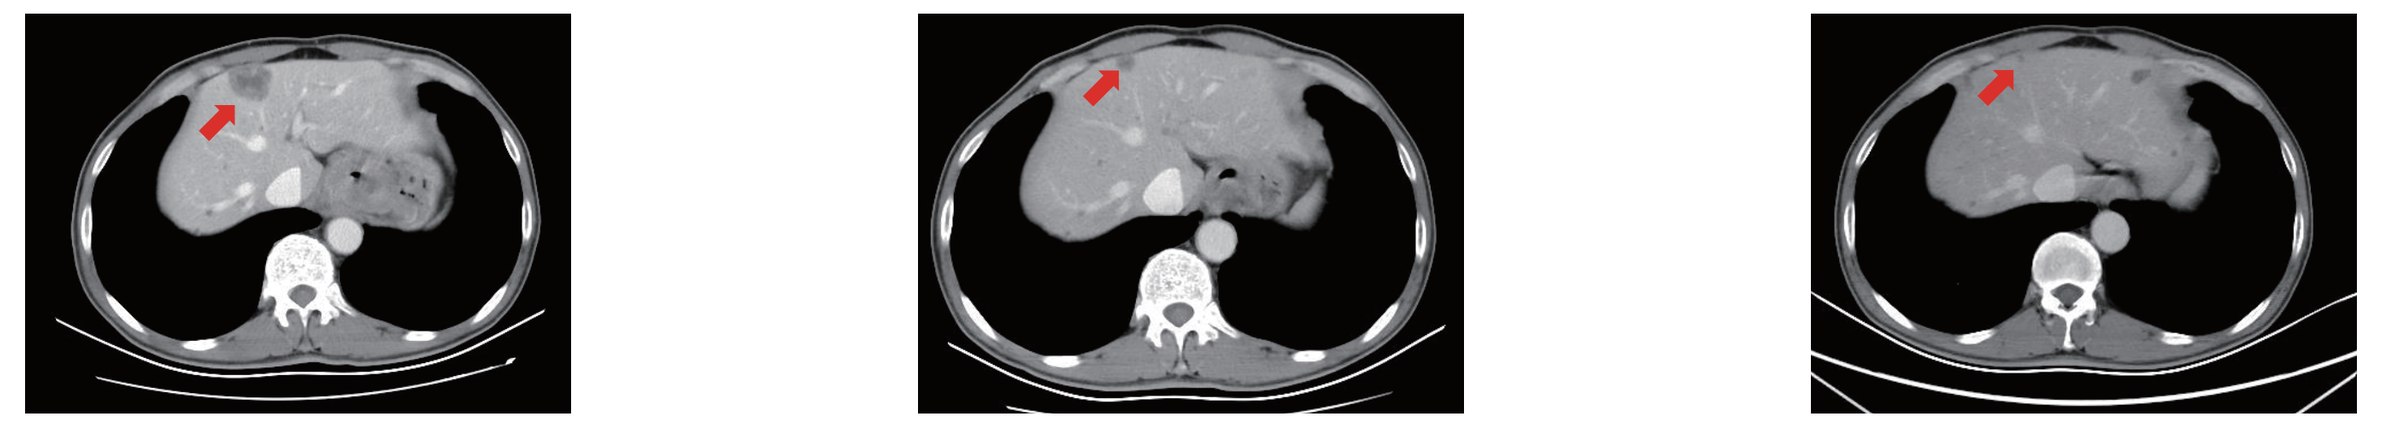

Baseline | **9 weeks, 63%↓ PR** | **18 weeks, 76%↓ PR**

Male, 58 years old, stage IV gastric cancer with liver metastasis; has completed 20 cycles of CS1001 treatment and 6 cycles of XELOX; remained on treatment as of 19 Feb 2020

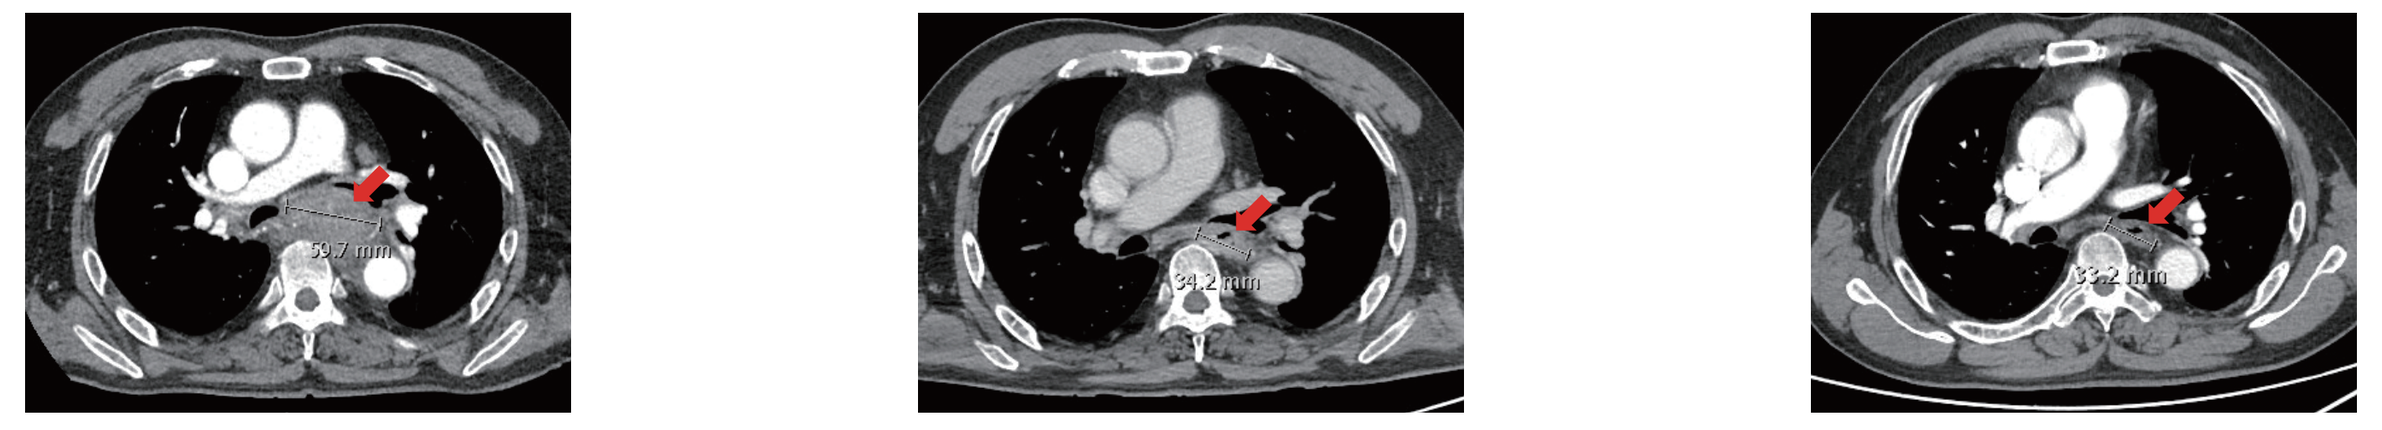

Baseline | **9 weeks, 43%↓ PR** | **18 weeks, 92%↓ PR**

Male, 67 years old, stage IV esophageal squamous cell carcinoma with regional lymph node metastasis; has completed 14 cycles of CS1001 treatment and 6 cycles of CF

Exposure and Safety (Safety Analysis Set):

- As of 19 Feb 2020, the median duration of CS1001 treatment was 232 (range: 21-523) and 172 (range: 21-488) days in GC/GEJ and ESCC cohorts, respectively
- In the GC/GEJ cohort, all-grade and Grade ≥ 3 AEs related to CS1001 occurred in 28 (96.6%) pts and 14 (48.3%) pts, respectively
 - The most common (n ≥ 2) Grade ≥ 3 CS1001-related AEs included platelet count decreased (n = 6), white blood cell count decreased (n = 3), neutrophil count decreased (n = 3), anaemia (n = 3), and fatigue (n = 2)
 - Three pts each had one CS1001-related AE leading to CS1001 discontinuation: Grade 3 hypothyroidism, Grade 3 abnormal liver function, and Grade 2 pneumonia
- In the ESCC cohort, all-grade and Grade ≥ 3 AEs related to CS1001 occurred in 34 (87.2%) pts and 16 (41.0%) pts, respectively
 - The most common (n ≥ 2) Grade ≥ 3 CS1001-related AEs included anaemia (n = 6), white blood cell count decreased (n = 3), neutrophil count decreased (n = 3), amylase increased (n = 3), platelet count decreased (n = 2), hyponatraemia (n = 2), and asthenia (n = 2)
 - Three pts each had one CS1001-related AE leading to CS1001 discontinuation: Grade 4 hyponatraemia, Grade 3 anaemia, and Grade 3 pneumonitis
 - One death with unknown cause was attributed to CS1001. The investigator was unable to exclude the causality as relevant information could not be obtained

Table 3 - Summary of Adverse Events (Safety Analysis Set)

	GC/GEJ (N = 29)	ESCC (N = 39)
Number of patients with at least one below event	n (%)	n (%)
TEAE	29 (100)	39 (100)
Grade 3/4/5 TEAE	20 (69.0)	34 (87.2)
TEAE related to CS1001	28 (96.6)	34 (87.2)
Grade 3/4/5 TEAE related CS1001	14 (48.3)	16 (41.0)
SAE	12 (41.4)	23 (59.0)
SAE related to CS1001	6 (20.7)	7 (17.9)
irAE	21 (72.4)	25 (64.1)
Grade 3/4 irAE	6 (20.7)	10 (25.6)
TEAE leading to CS1001 discontinuation	4 (13.8)	5 (12.8)
TEAE leading to discontinuation of CS1001 or chemotherapy	8 (27.6)	8 (20.5)
TEAE leading to CS1001 dose interruption	1 (3.4)	1 (2.6)
TEAE leading to treatment cycle delay	16 (55.2)	19 (48.7)
TEAE leading to death	0	3 (7.7)
Infusion-related reaction	2 (6.9)	3 (7.7)

TEAE – Treatment Emergent Adverse Event; SAE – Serious Adverse Event; irAE – Immune-related TEAE

CONCLUSIONS

- CS1001 in combination with standard-of-care (SoC) chemotherapy demonstrated robust and durable anti-tumor activities with a tolerable safety profile in first-line treatment setting among patients with advanced GC/GEJ and ESCC
 - GC/GEJ – ORR 62%, mDoR 11.3 months, mPFS 8.3 months, mOS 17.0 months
 - ESCC - ORR 68%, mDoR not reached, mPFS 9.0 months, mOS not reached
- Current data support further development of CS1001 plus SoC chemotherapy in advanced GC/GEJ and ESCC. Two double-blinded, randomized phase 3 studies are ongoing in China:
 - GEMSTONE-303 (CS1001-303, NCT03802591) investigating CS1001 in combination with XELOX in patients with advanced GC/GEJ
 - GEMSTONE-304 (CS1001-304, NCT04187352) investigating CS1001 in combination with CF in patients with advanced ESCC

Acknowledgement

We thank all the patients who participated in this study, their families, participating investigators and clinical sites. This study is sponsored by CStone Pharmaceuticals (Su Zhou) Co., Ltd. Medical writing assistance was provided by Qinzhou Qi from CStone Pharmaceuticals.

Disclosure

All authors have declared no conflicts of interest.

Poster presented at ESMO Virtual Congress 2020, Science Weekend, 19-21 Sept 2020

Contact corresponding author: ClinicalDevelopment@cstonepharma.com

ClinicalTrials.gov identifier: NCT03312842

QR Code

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

基石药业 CSTONE PHARMACEUTICALS