Phase 1 Study of Fisogatinib (BLU-554) in Patients (pts) with Advanced Hepatocellular Carcinoma (aHCC) Expressing FGF19: Preliminary Results from Tyrosine Kinase Inhibitor (TKI)-naïve Chinese Pts in Part 3 of the Study

Abstract 7842

Shukui Qin[1], Bo Li[2], Jufeng Wang[3], Aibing Xu[4], Yabing Guo[5], Weijia Fang[6], Ying Cheng[7], Christopher D. Turner[8], Yan Wu[9], Xin Chen[9], Jin Hu[9], Yu Sun[9], Yedong Wang[9], Archie N.Tse[9], Jason Yang[9]

1. Qinhuai Medical District, General Hospital of Eastern Theater Command, Nanjing, China; 2. West China Hospital of Sichuan University, Chengdu, China; 3. Affiliated Cancer Hospital of Zhengzhou University, Henan Cancer Hospital, Zhengzhou, China; 4. Nantong Tumor Hospital, Nantong, China; 5. Nanfang Hospital, Guangzhou, China; 6. The First Affiliated Hospital, School of Medicine, Zhejiang University, Hangzhou, China; 7. Jilin Cancer Hospital, Changchun, China; 8. Blueprint Medicines Corporation, Cambridge, United States; 9. CStone Pharmaceuticals (Suzhou) Co., Ltd., Suzhou, China

BACKGROUND

Emerging data have implicated aberrant FGF19-FGFR4 signaling pathway as an oncogenic driver in HCC. Fisogatinib, a highly potent and selective FGFR4 inhibitor, was first evaluated in the global phase 1 BLU-554-1101 study in aHCC pts (NCT02508467). The study consists of 3 parts. Part 1 consists of dose escalation employing a 3+3 design to define the maximum tolerated dose/recommended Phase 2 dose (MTD/RP2D); Part 2 consists of dose expansion exploring the MTD/RP2D for the QD dosing schedule. Parts 1 and 2 of the study demonstrated that FGFR4 inhibition was tolerable and clinically active in HCC expressing FGF19. Further investigation of fisogatinib in FGF19-positive, tyrosine kinase inhibitor (TKI) treatment-naïve pts with aHCC (part 3) is ongoing globally, and we report preliminary data from Chinese pts.

Figure 1 FGF19-FGFR4 Pathway [1-7]

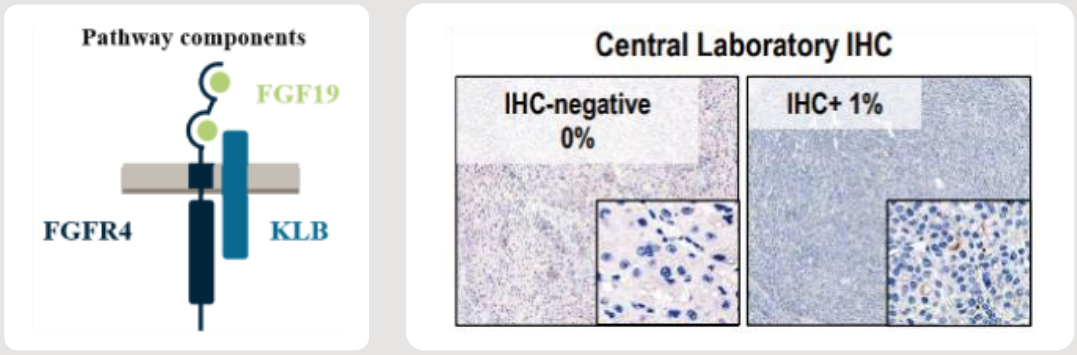
Pathway components
FGF19
FGFR4
KLB
Central Laboratory IHC
IHC-negative 0%
IHC+ 1%

- Aberrant FGF19-FGFR4 signaling identified as a potential HCC driver
- FGF19 is a mitogen that signals via FGFR4 and KLB
- Normal liver and HCC consistently express FGFR4 and KLB; but only HCC aberrantly expresses FGF19 ligand (27% were positive for FGF19 staining ≥ 1%)[7]
- Aberrant FGF19 expression may drive HCC and confer a poor prognosis

STUDY DESIGN

Figure 2 QD Dosing Schedule Study Schematic

Part 1: Dose Escalation, QD schedule
- 3+3 Design
- N = 25

Part 2: Expansion (600mg QD) QD schedule
- N = 81
- Prospective enrollment via IHC screening at central laboratory
- Retrospective stratification via central laboratory FISH

Part 3: Expansion (FGF19 IHC+, TKI-naïve) fisogatinib 600mg QD
- N = 31 (Chinese: 11)
- IHC ≥ 1%
- Prospective enrollment via IHC screening at central laboratory

Primary Endpoint:
- Safety

Secondary Endpoints:
- Pharmacokinetics
- FGF19 protein levels
- Objective Response Rate (ORR), Duration of Response (DOR), Disease Control Rate (DCR), Progression-free survival (PFS), as per RECIST, version 1.1 by investigator assessment
- Overall survival (OS)

Key Inclusion Criteria (Part 3):
- The patient must have FGF19 IHC+ HCC
- The patient must not receive prior treatment with a TKI
- The patient must have at least one measurable lesion evaluable by RECIST, version 1.1
- Eastern Cooperative Oncology Group (ECOG) performance status of 0-1

RESULTS

Demographic and Baseline Characteristics
- As of 30 Apr 2020, 11 Chinese pts were treated; 5 pts remained on treatment and 6 pts discontinued due to disease progression

Table 1 Demographic and Baseline Characteristics

	Part 3, China (N=11)
Age, years (median, range)	58 (27-72)
Age, n (%)	
<65 years	9 (81.8)
≥65 years	2 (18.2)
Sex	
Male	8 (72.7)
Female	3 (27.3)
ECOG PS	
0	5 (45.5)
1	6 (54.5)
BCLC (Barcelona Clinic Liver Cancer) stage	
A	1 (9.1)
B	1 (9.1)
C	9 (81.8)
Etiology	
Hepatitis B	11 (100.0)
MVI (macroscopic vascular invasion)	
Yes	5 (45.5)
No	6 (54.5)
Extrahepatic metastasis	
Yes	7 (63.6)
No	4 (36.4)
Prior systemic anti-cancer therapy	
No	8 (72.7)
Immune oncology therapy	1 (9.1)
Chemotherapy	2 (18.2)

Preliminary Efficacy Data
- All 11 pts were included in efficacy analysis. 4 pts (36.4%) reached partial response, 3 responses were confirmed; 4 had stable disease. The disease control rate was 72.7%
- There was no direct correlation between FGF19 IHC levels and clinical response; among the 4 responders, the FGF19 IHC levels were between 5% and 100%

Table 2 Best Overall Response in FGF19 IHC+

	Part 3, China (N=11) n (%)	95% CI
ORR (CR+PR)	4 (36.4%)*	[10.9%, 69.2%]
Complete response (CR)	0	
Partial response (PR)	4 (36.4%)*	[10.9%, 69.2%]
Stable disease (SD)	4 (36.4%)	[10.9%, 69.2%]
Progressive disease (PD)	3 (27.3%)	[6.0%, 61.0%]
DCR (CR+PR+SD)	8 (72.7%)	[39.0%, 94.0%]

* 3 PRs were confirmed

Figure 3 Swimmer Plot of Treatment Durations, Best Overall Response and Progression with not Confirmed CR or PR

3 responses lasted for > 12 weeks and are still ongoing

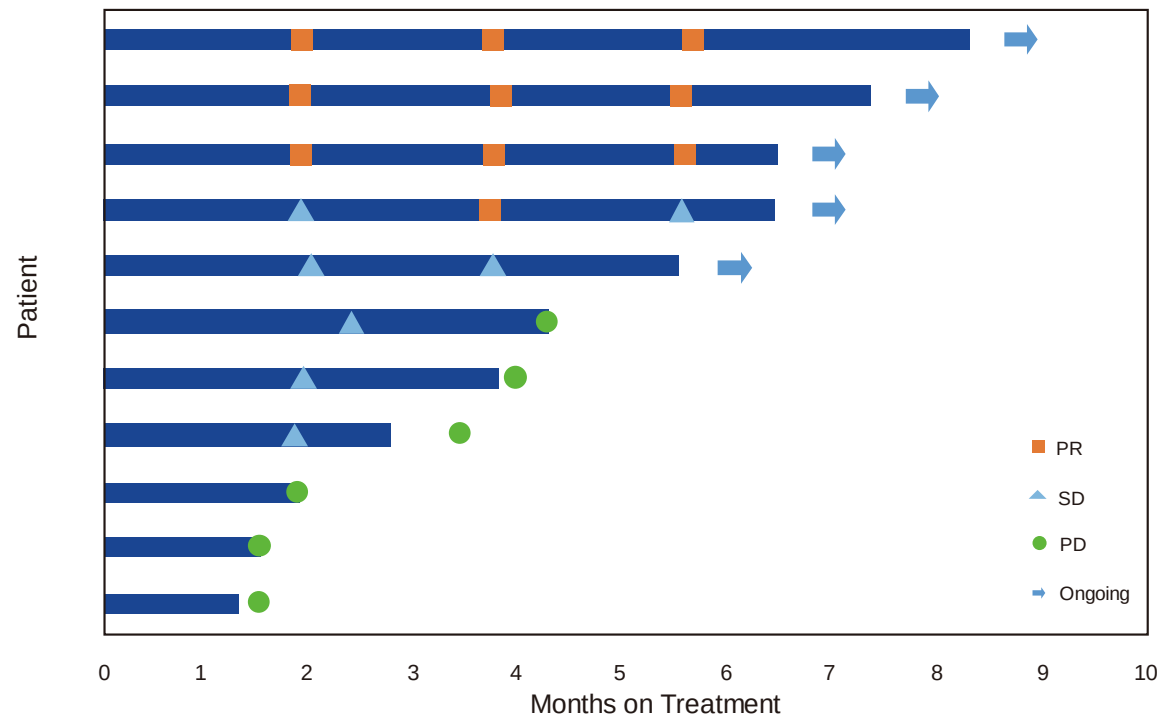
Patient
PR
SD
PD
Ongoing
0
1
2
3
4
5
6
7
8
9
10
Months on Treatment

Figure 4 Best Percentage Change from Baseline in Sum of Products of Diameters

3 pts had the max tumor reduction > 60%

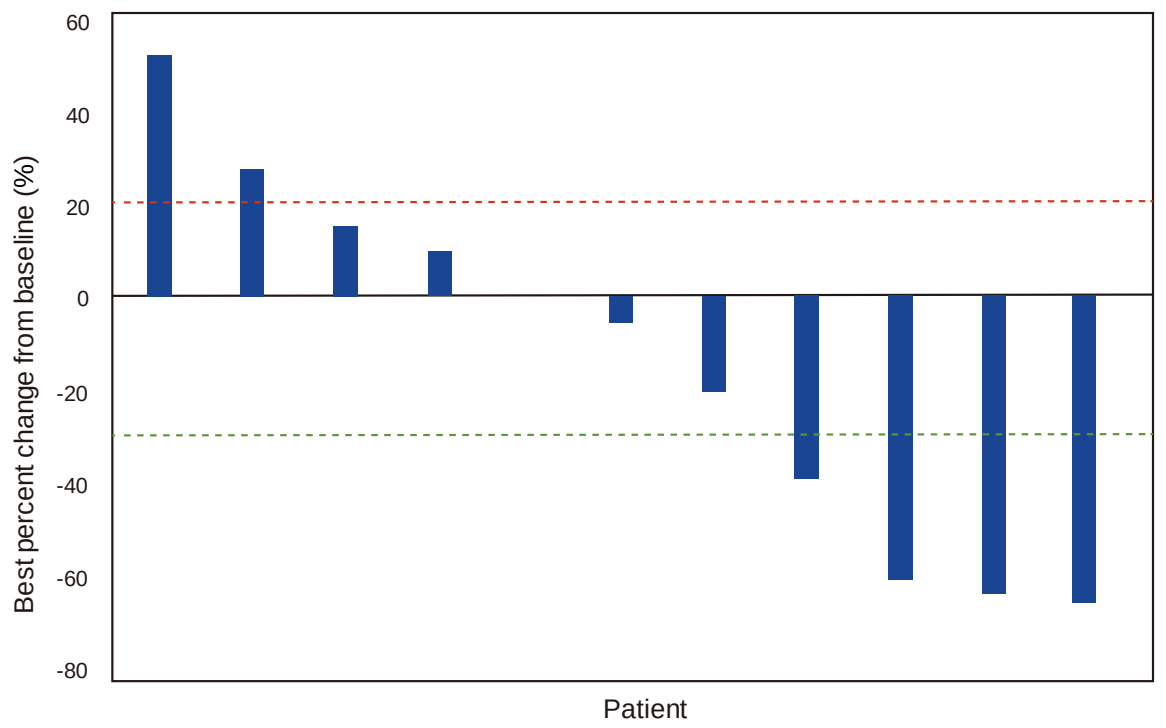
60
40
20
0
-20
-40
-60
-80
Best percent change from baseline (%)
Patient

Safety Data
- As of 30 Apr 2020, all pts had treatment-related adverse events (TRAEs), with the most common ones being ALT increased (90.9%), diarrhoea (81.8%), AST increased (72.7%) and blood bilirubin increased (63.6%)
- 3 (27.3%) pts had Grade 3 TRAEs
- No Grade 4/5 TRAEs occurred
- All TRAEs were manageable
- No AE leading to treatment discontinuation
- One treatment-related SAE was Grade 3 abnormal liver function

Table 3 TRAEs in ≥10% of Chinese Pts Treated with 600mg QD Dosing

Preferred Term	Part 3, China (N=11) Any Grade n (%)	≥ Grade 3 n (%)
Number of pts with at least one event	11 (100.0)	3 (27.3)
ALT increased	10 (90.9)	1 (9.1)
Diarrhoea	9 (81.8)	0
AST increased	8 (72.7)	1 (9.1)
Blood bilirubin increased	7 (63.6)	1 (9.1)
Platelet count decreased	6 (54.6)	0
Nausea	4 (36.4)	0
Blood creatinine increased	2 (18.2)	0
Dyspepsia	2 (18.2)	0
Headache	2 (18.2)	0
White blood cell count decreased	2 (18.2)	0

CONCLUSIONS

- Fisogatinib demonstrated a manageable side effect profile in Chinese pts that is consistent with FGFR4 pathway inhibition
- Encouraging preliminary efficacy was observed in FGF19-positive, TKI treatment-naïve Chinese pts, which further validated the oncogenic driver role of the FGFR4 pathway and the use of FGF19 as a biomarker for patient selection in HCC
- A clinical trial to evaluate fisogatinib in combination with CS1001 (anti-PD-L1 mAb) in aHCC is ongoing

References

1. Llovet JM et al (2016) Nature Reviews Disease Primers 2: 1–23;
2. Miura S et al (2012) BMC Cancer 12:56;
3. Hyeon J et al (2013) Dig Dis Sci 58:1916-1922;
4. Schulze K et al (2015) Nature Genetics 47:505–511;
5. Hatlen MA et al (2019) Cancer Discov 9:1689;
6. Kim RD et al, presented on 2017 ESMO Abs 365O;
7. Kim RD et al, Cancer Discov. 2019 Dec;9(12):1696-1707.

Acknowledgements

We would like to thank the pts for their participation, their families, and the investigators and sites participated in this study. This study is sponsored by CStone Pharmaceuticals (Suzhou), Co., Ltd. Medical writing assistance was provided by Vivi Meng from CStone Pharmaceuticals.

ClinicalTrial.gov numbers NCT02508467

QR CODE

Scan to visit CStone Virtual Booth and e-poster. Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

virtual booth E-poster

The 23rd Chinese Society of Clinical Oncology 2020.9.19-9.26